LIPIDATED PEPTIDE HYDROGELS: A PLATFORM FOR THE DELIVERY OF THERAPEUTICS FOR THE TREATMENT OF INFLAMMATORY BOWEL DISEASES

__Marilisa Pia Dimmito__[1], Antonio Di Stefano[1,2], Lisa Marinelli[1,2], Annalisa Di Rienzo[1], Giorgia Carraro[3], Barbara Albertini[4], Alessandra Rapino[4], Eleonora Chiara Toto[1], Ivana Cacciatore[1,2]

[1]University "G. D'Annunzio of Chieti-Pescara, Department of Pharmacy, Via Dei Vestini 31, 66100, Chieti, Italy;
[2]Algo Biotechnologies S.R.L., University of Chieti-Pescara, Via dei Vestini 1, 66100, Chieti, Italy;
[3]Department of Pharmaceutical and Pharmacological Sciences, University of Padova, Via F. Marzolo 5, 35131, Padova, Italy
[4]Dompé Pharmaceutical, Via Campo di Pile, Nucleo Industriale Pile, 67100, L'Aquila, Italy
marilisa.dimmito@unich.it

INTRODUCTION

Antimicrobial peptides (AMPs) are widely distributed through the plant and animal kingdoms playing a central role in the evolution of superior organisms. AMPs implication in the treatment of infectious diseases is highly recognized, and, unlike commercially available antibacterial drugs readily circumvented by bacteria, the resistance to them is surprisingly doubtful [1]. AMPs can be efficacious in eosinophilic esophagitis and inflammatory bowel diseases (IBD), often provoked by intestinal microbiota imbalance, whose main symptoms include inflammation, severe pain, loss of barrier functions, and susceptibility to infections [2]. A small peptide with sequence Ser-Asn-Ala (SNA) showed moderate antibacterial activity against both Gram-positive and negative bacteria [3]. However, SNA like other small AMPs, has shortcomings such as enzymatic degradation, physical and chemical instabilities, and short half-life, hampering its applicability as therapeutics. To overcome these drawbacks, in this work, we reported the lipidation of a series of SNA analogues, obtained by using different combinations of D-, and L-aminoacids. On one hand, this strategy is aimed to increase the stability as well as the antimicrobial activity of SNA, on the other hand, to obtain self-assembly lipo-peptides able to arrange into ordered supramolecular structures (hydrogels) as a platform for drug delivery [4].

LIPO-PEPTIDES PREPARATION

Lipo-derivatives of SNA (**MPD_02-09**) have been prepared by standard solution phase peptide synthesis and were used as starting material to produce supramolecular hydrogels (**MPDh_02-09**). The procedure is reported in **Scheme 1**.

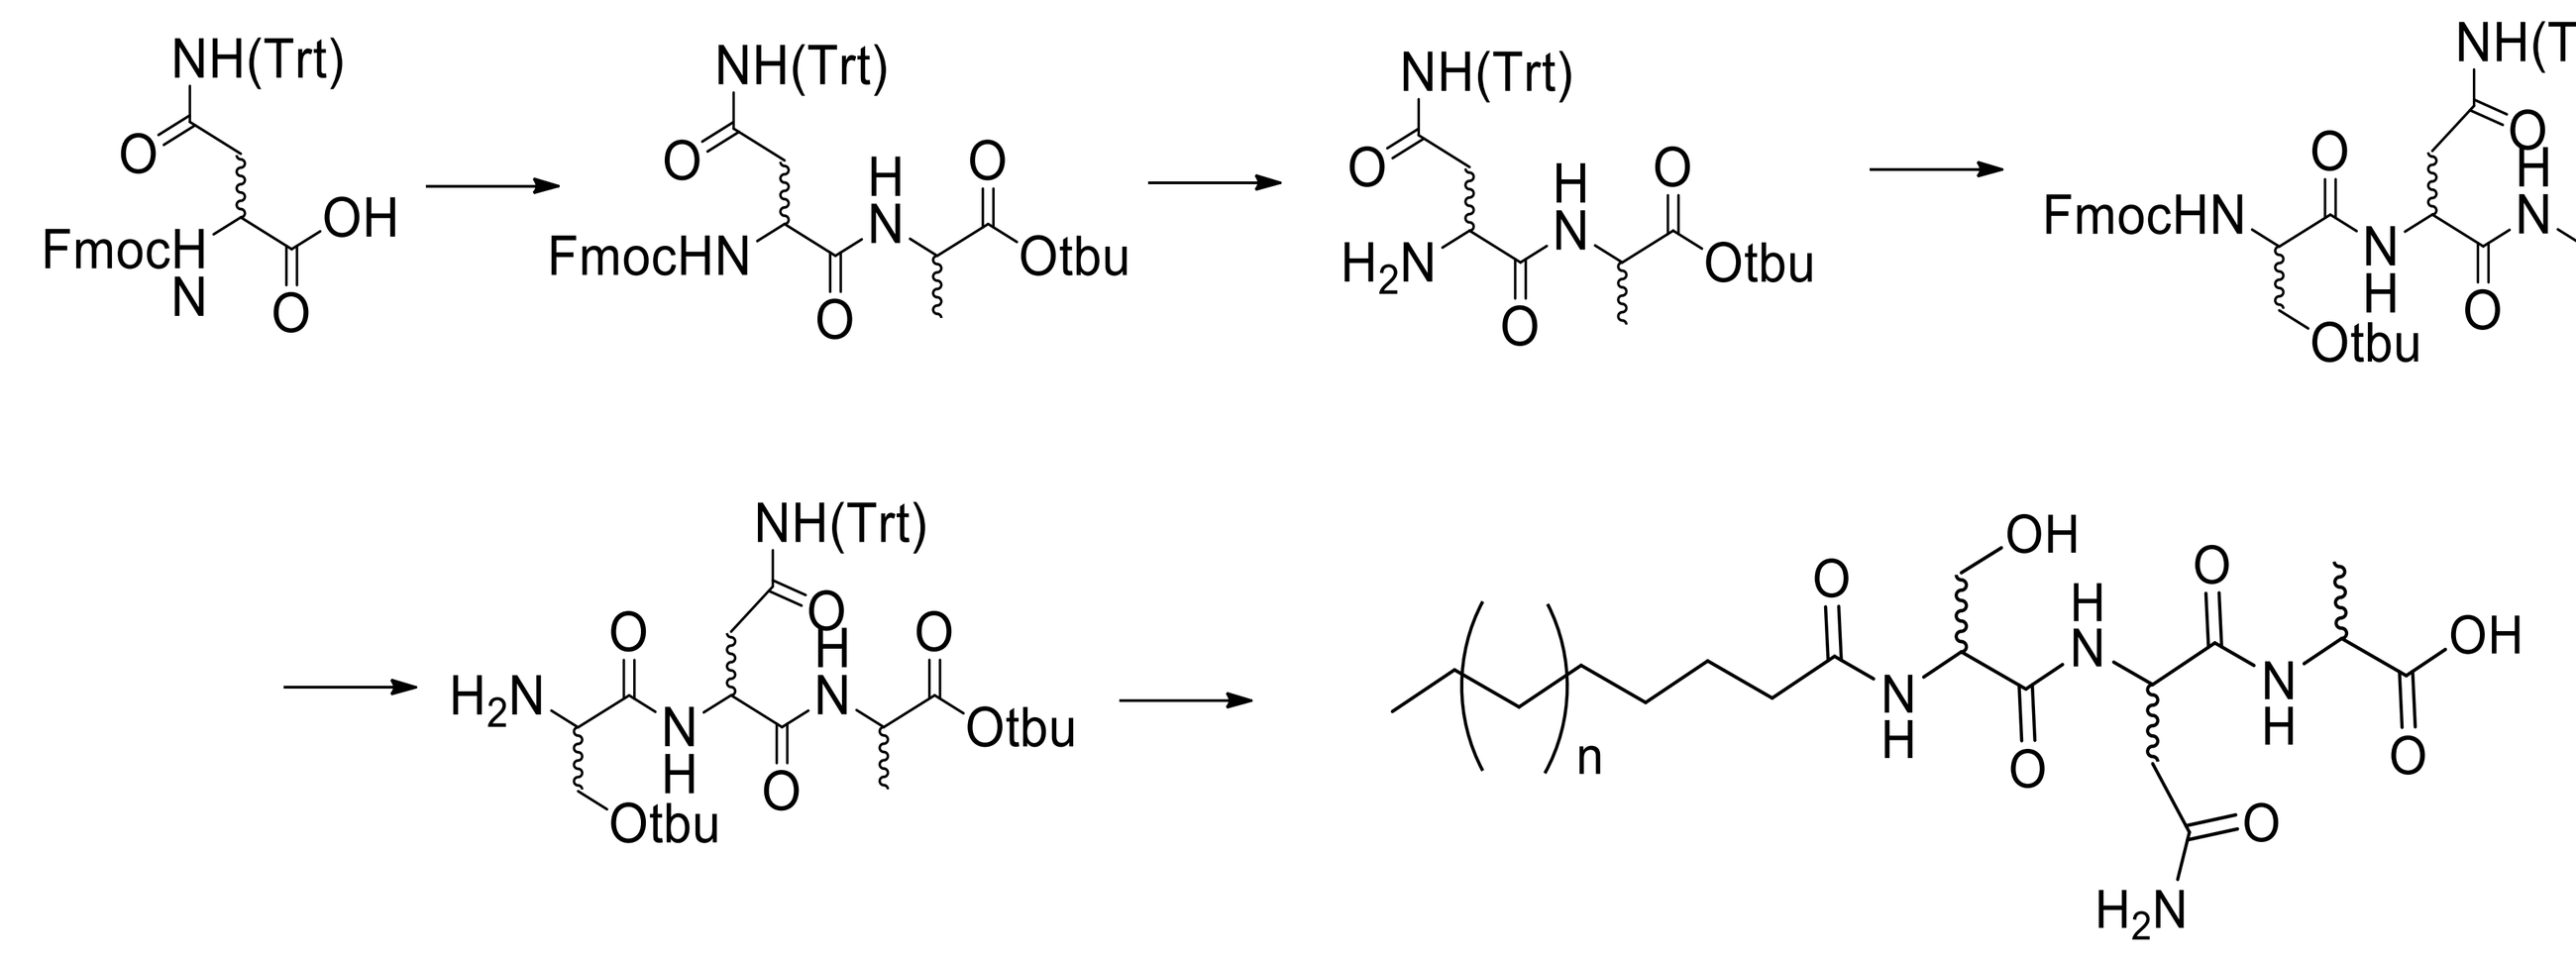

NH(Trt)
FmocHN
OH
Otbu
H2N

MPD_02-09

Scheme 1. General preparation scheme for lipo-peptides **MPD_02-09**.

LIPO-PEPTIDE-BASED HYDROGEL PREPARATION

MPD_02-09 hydrogels were prepared by dispersing the exact amounts of lipo-peptides to obtain 4 and 8mM final concentrations in PBS (150 mM, pH 7.4) and heating to 80 °C allowing the solubilization. Hydrogels with CGC from 4-8 mM and surface tension ranging from 46 to 65 mN/m were obtained after cooling down to room temperature (**Fig. 1**).

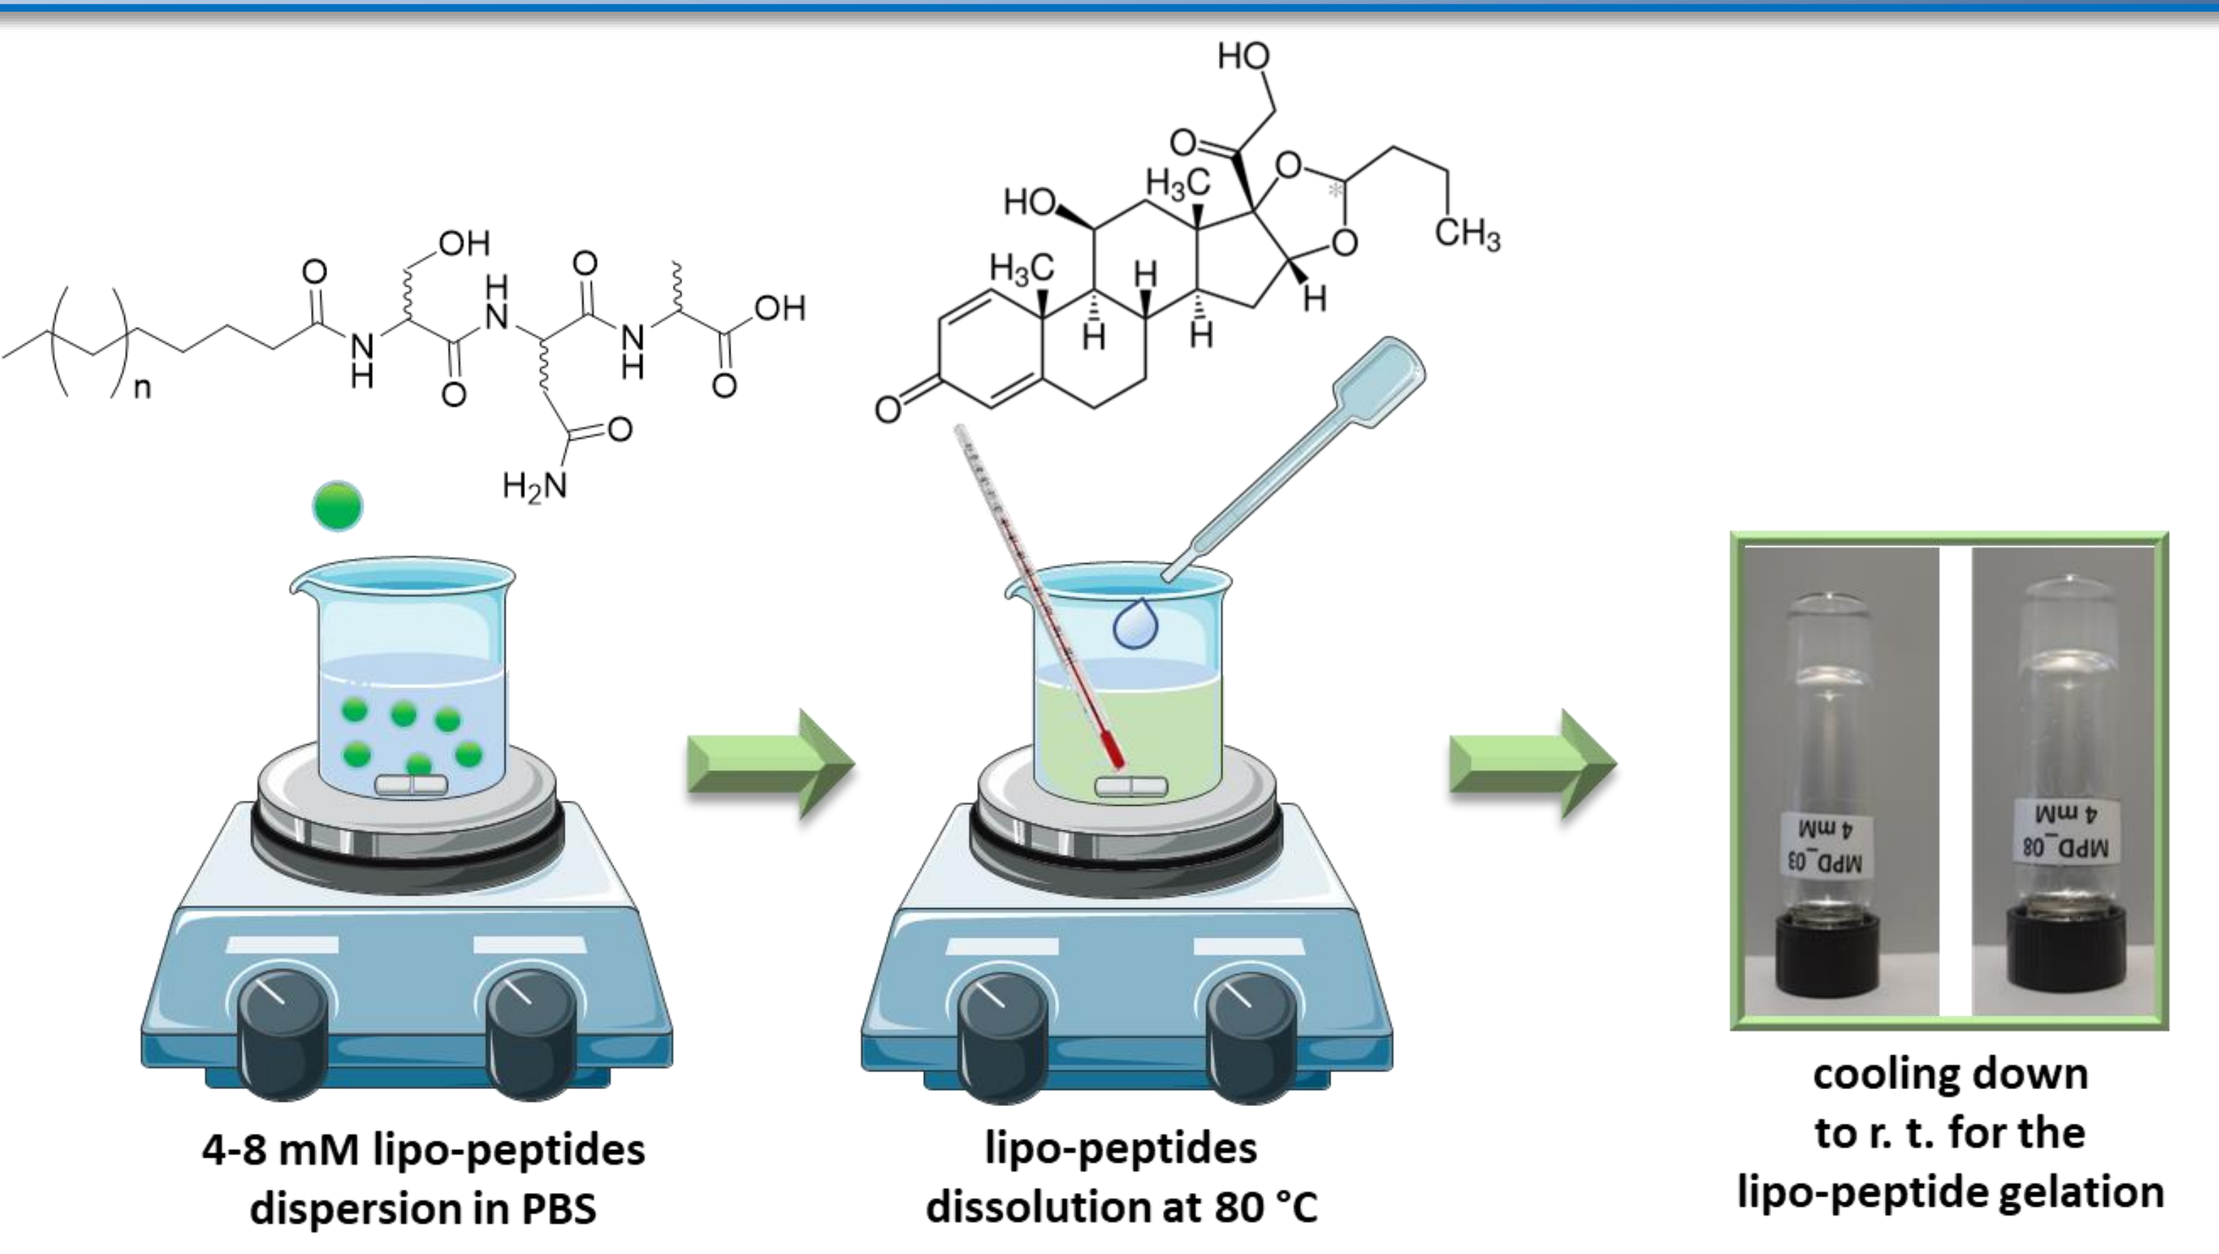

4-8 mM lipo-peptides dispersion in PBS
lipo-peptides dissolution at 80 °C
cooling down to r. t. for the lipo-peptide gelation

Fig. 1. MPD*h* preparation.

RHEOLOGICAL PROPERTIES

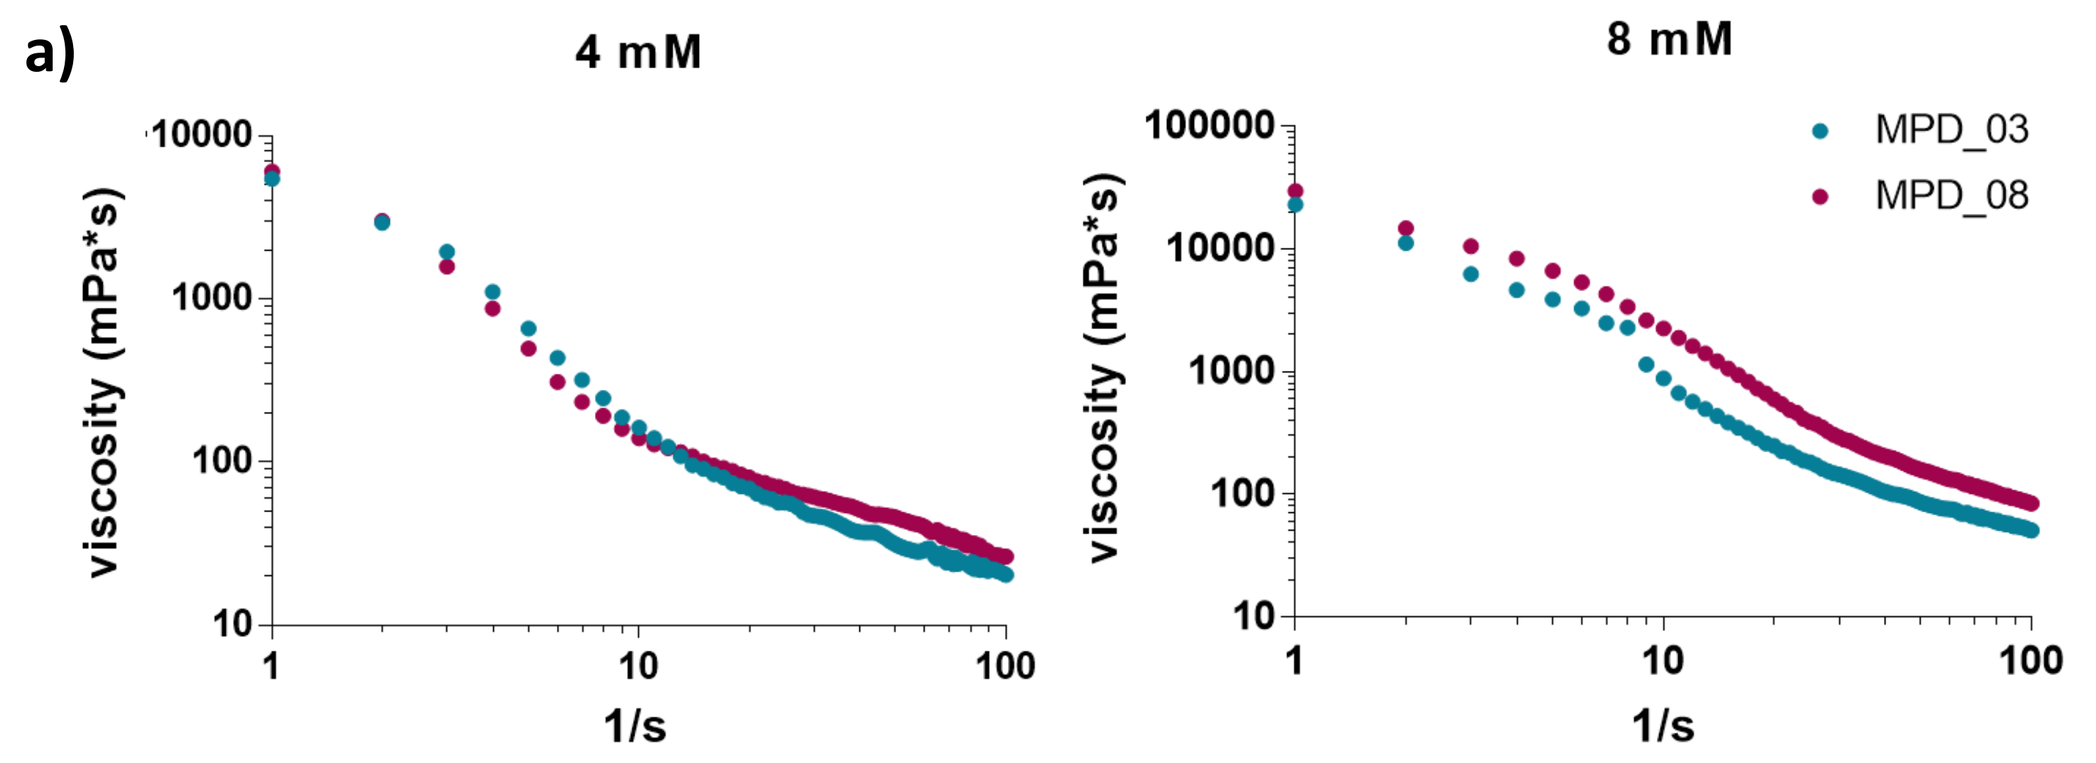

a)
4 mM
8 mM
viscosity (mPa*s)
1/s
MPD_03
MPD_08

Fig. 2. Flow curve of **MPD_03** and **MPD_08** unloaded hydrogels.

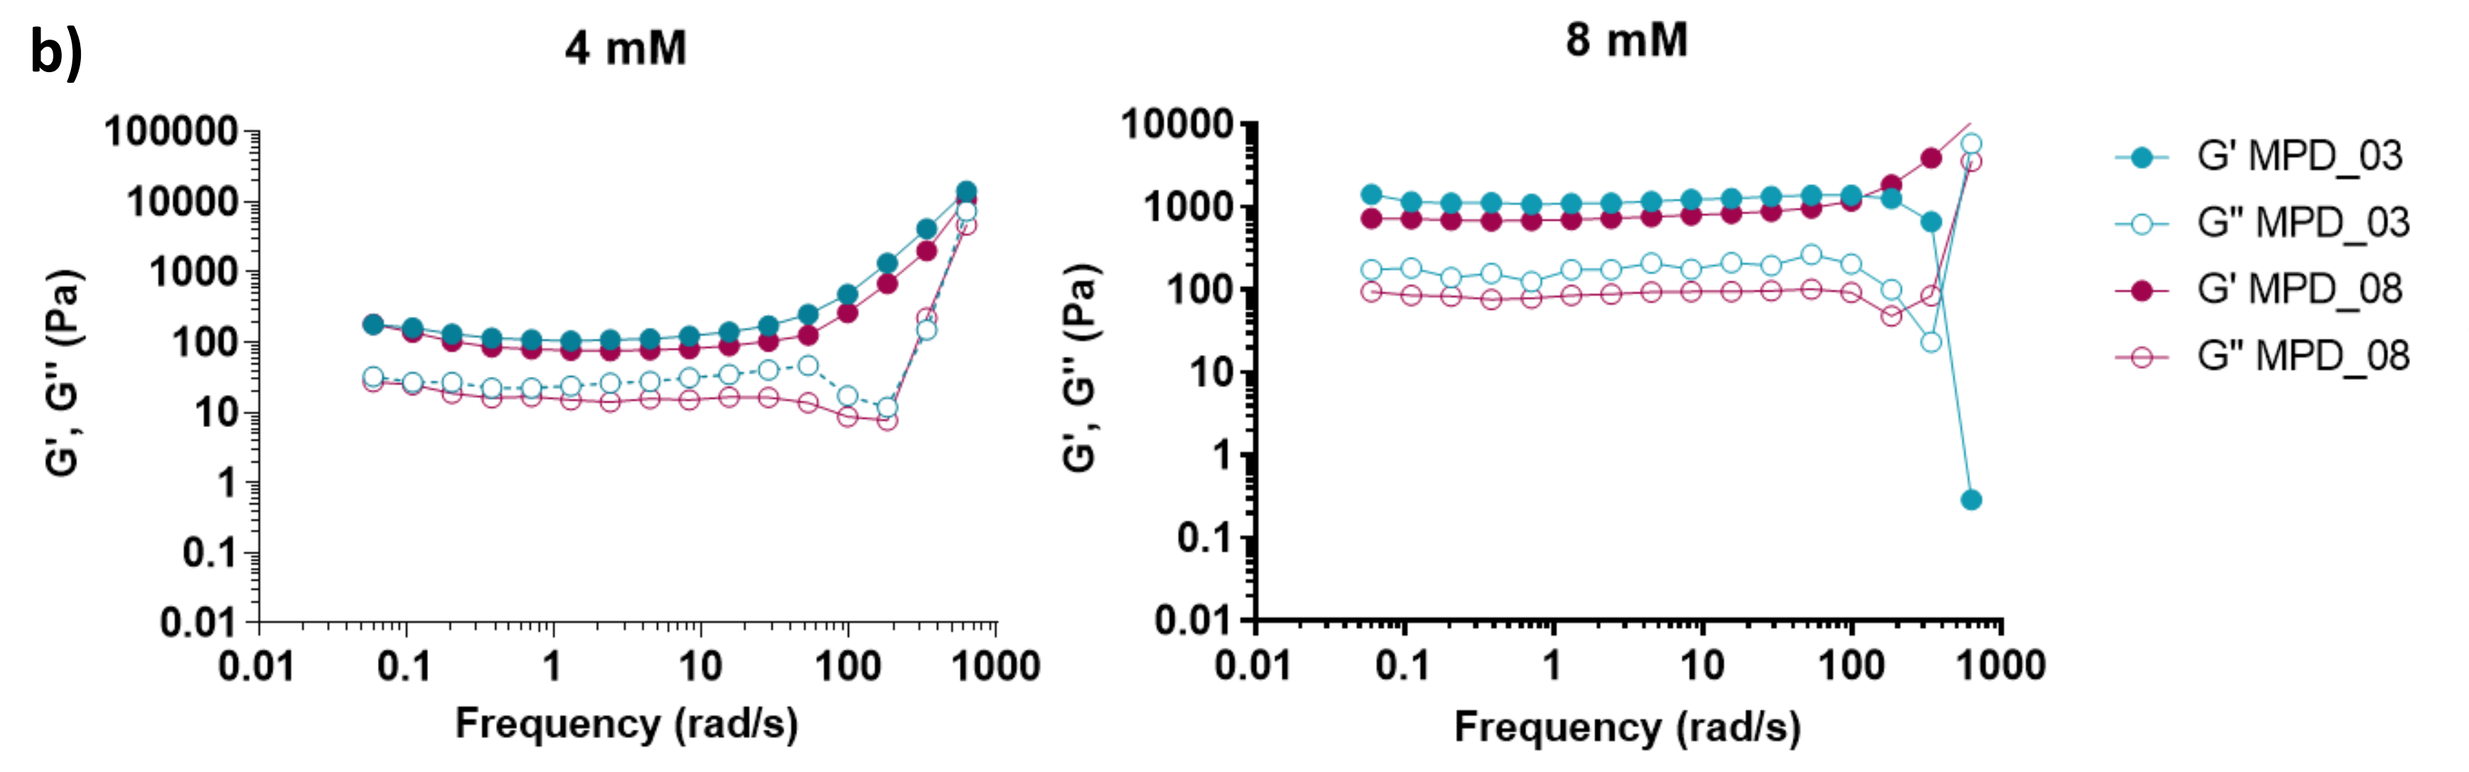

b)
4 mM
8 mM
G', G'' (Pa)
Frequency (rad/s)
G' MPD_03
G" MPD_03
G' MPD_08
G" MPD_08

Fig. 3. G' and G'' moduli of **MPD_03** and **MPD_08** unloaded hydrogels.

Fig. 2. **a**) and **b**) shows shear-thinning behavior in the range of considered shear rates. Additionally, the solid-like behavior was confirmed. As reported in **Fig. 3. a**) and **b**) a solid-like behavior with viscoelasticity and stiffness strongly dependent on the concentration with the storage modulus G' higher than the loss modulus G". The constant plateau region describe the strength of 3D network of the self-assembled material.

MORPHOLOGICAL ANALYSIS

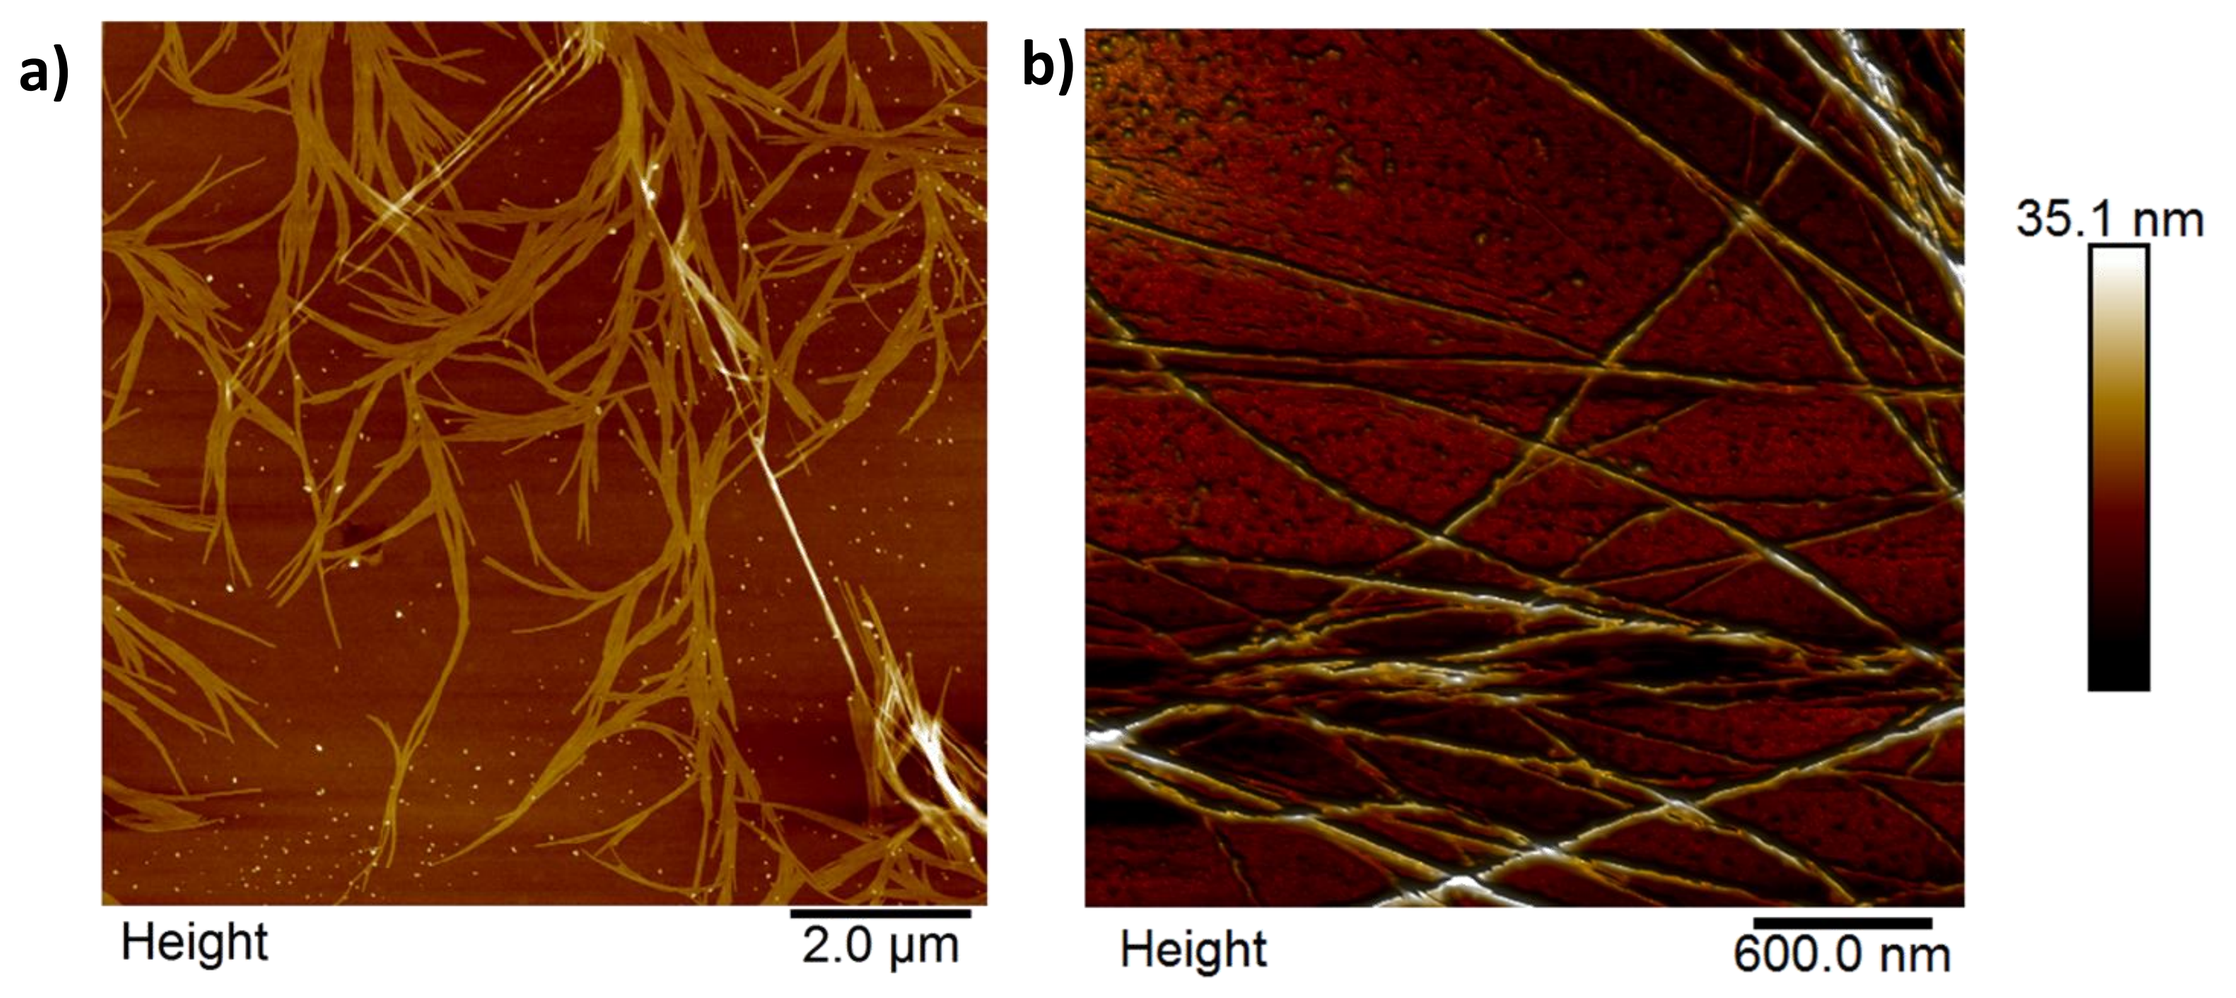

a)
b)
35.1 nm
Height
2.0 µm
Height
600.0 nm

Fig. 4. AFM images of **MPD_03 a)** and **MPD_08 b).**

To characterize **MPD_02-09** morphology, AFM analysis have been carried out. It was found that 4-8 mM lipo-peptides formed long nanofibers with an average diameter from 73 to 120 nm.

In **Fig. 4** the AFM morphology of **MPD_03** and **MPD_08** is shown.

IN VITRO RELEASE STUDIES

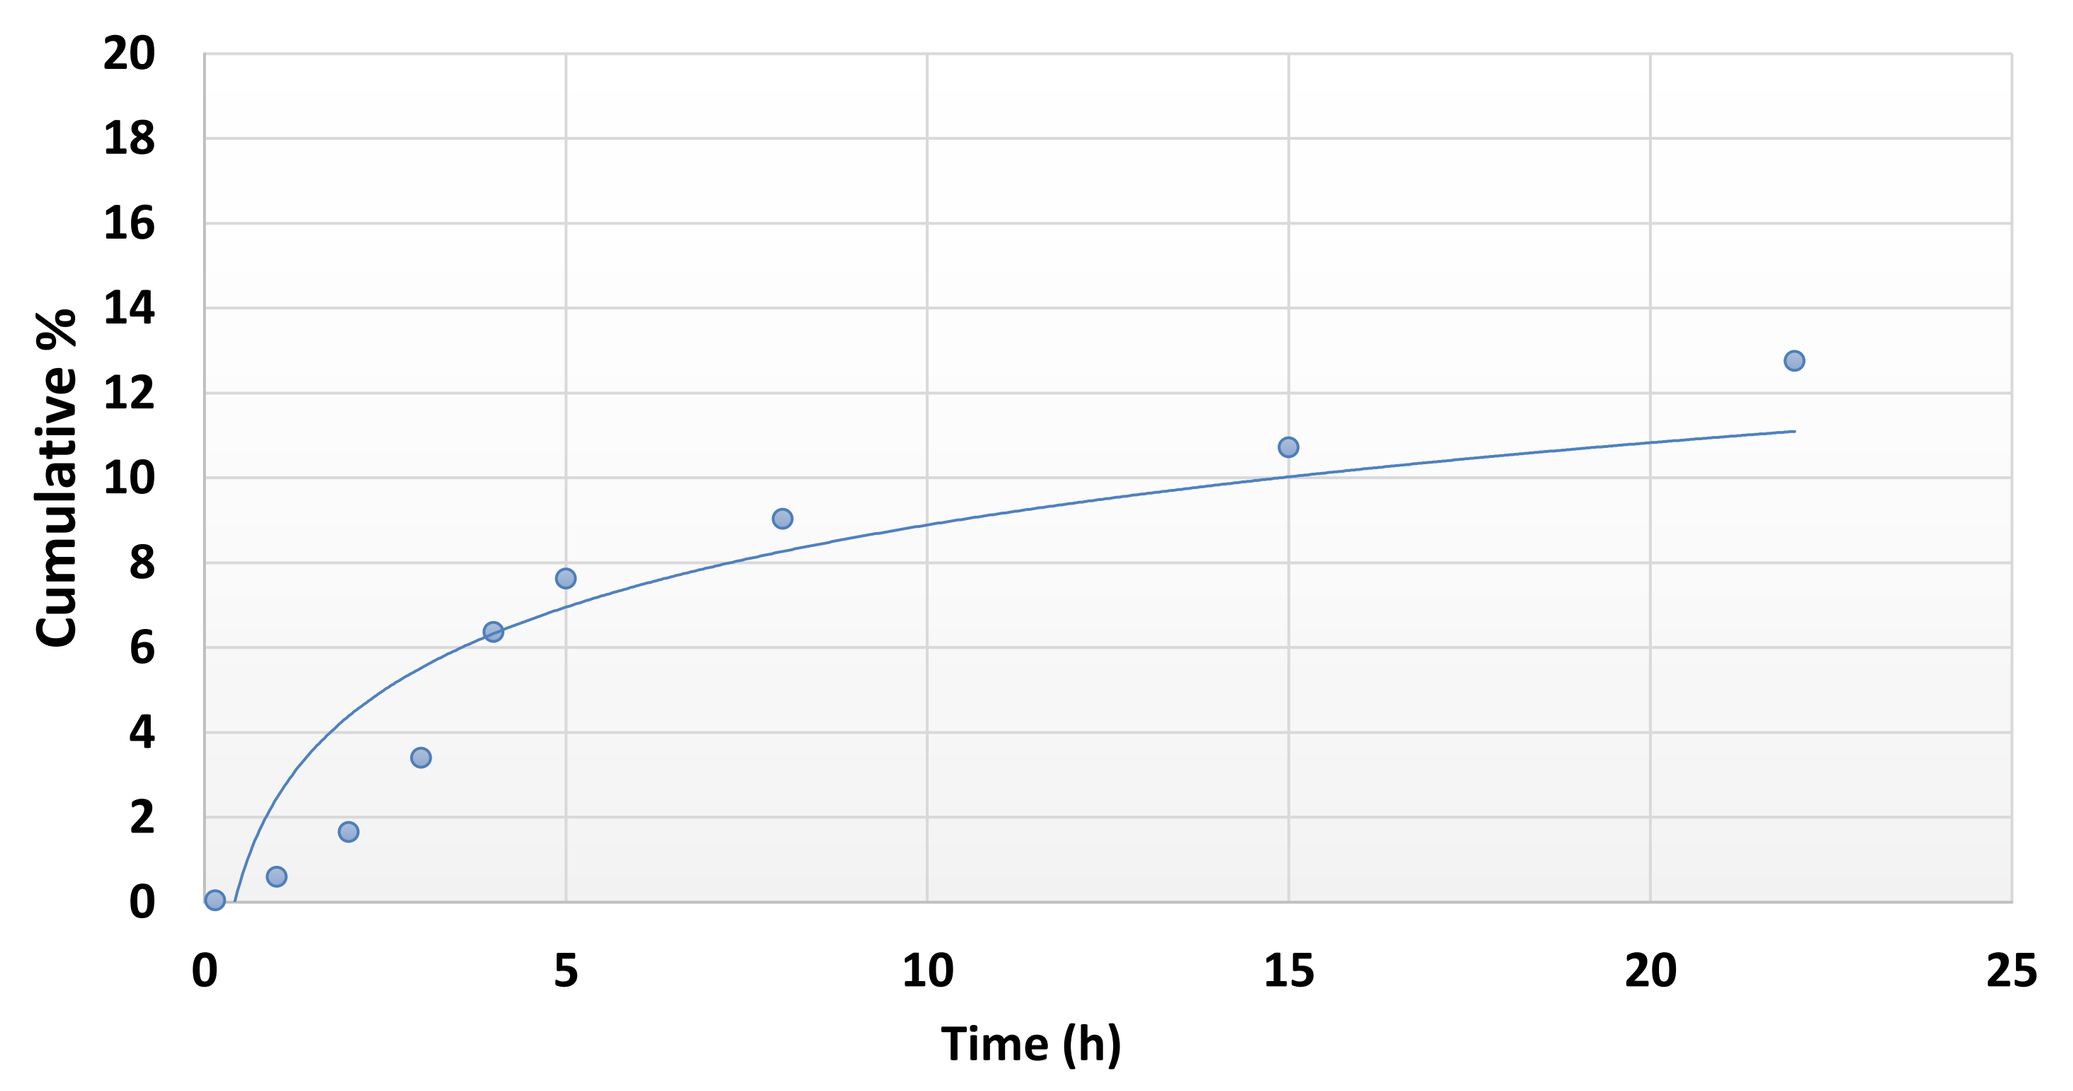

Cumulative %
Time (h)

Fig. 5. Release profile of budesonide in 150 mM PBS buffer pH 7.4, 25 % ethanol.

In vitro release studies were performed in using a Franz Cell device equipped with a cellulose membrane (*cut-off* 12400 Da) in 150 mM PBS buffer pH 7.4 enriched with 25% Ethanol at 37 ± 2°C. The result indicates an initial small burst release followed by a continuous release which provides approximately 13% of the entrapped budesonide as shown in **Fig. 5**. These results suggest that drug-loaded MPDs hydrogel provides a continuous release within 24 h.

IN VITRO CITOTOXICITY AND BIOLOGICAL ASSAYS

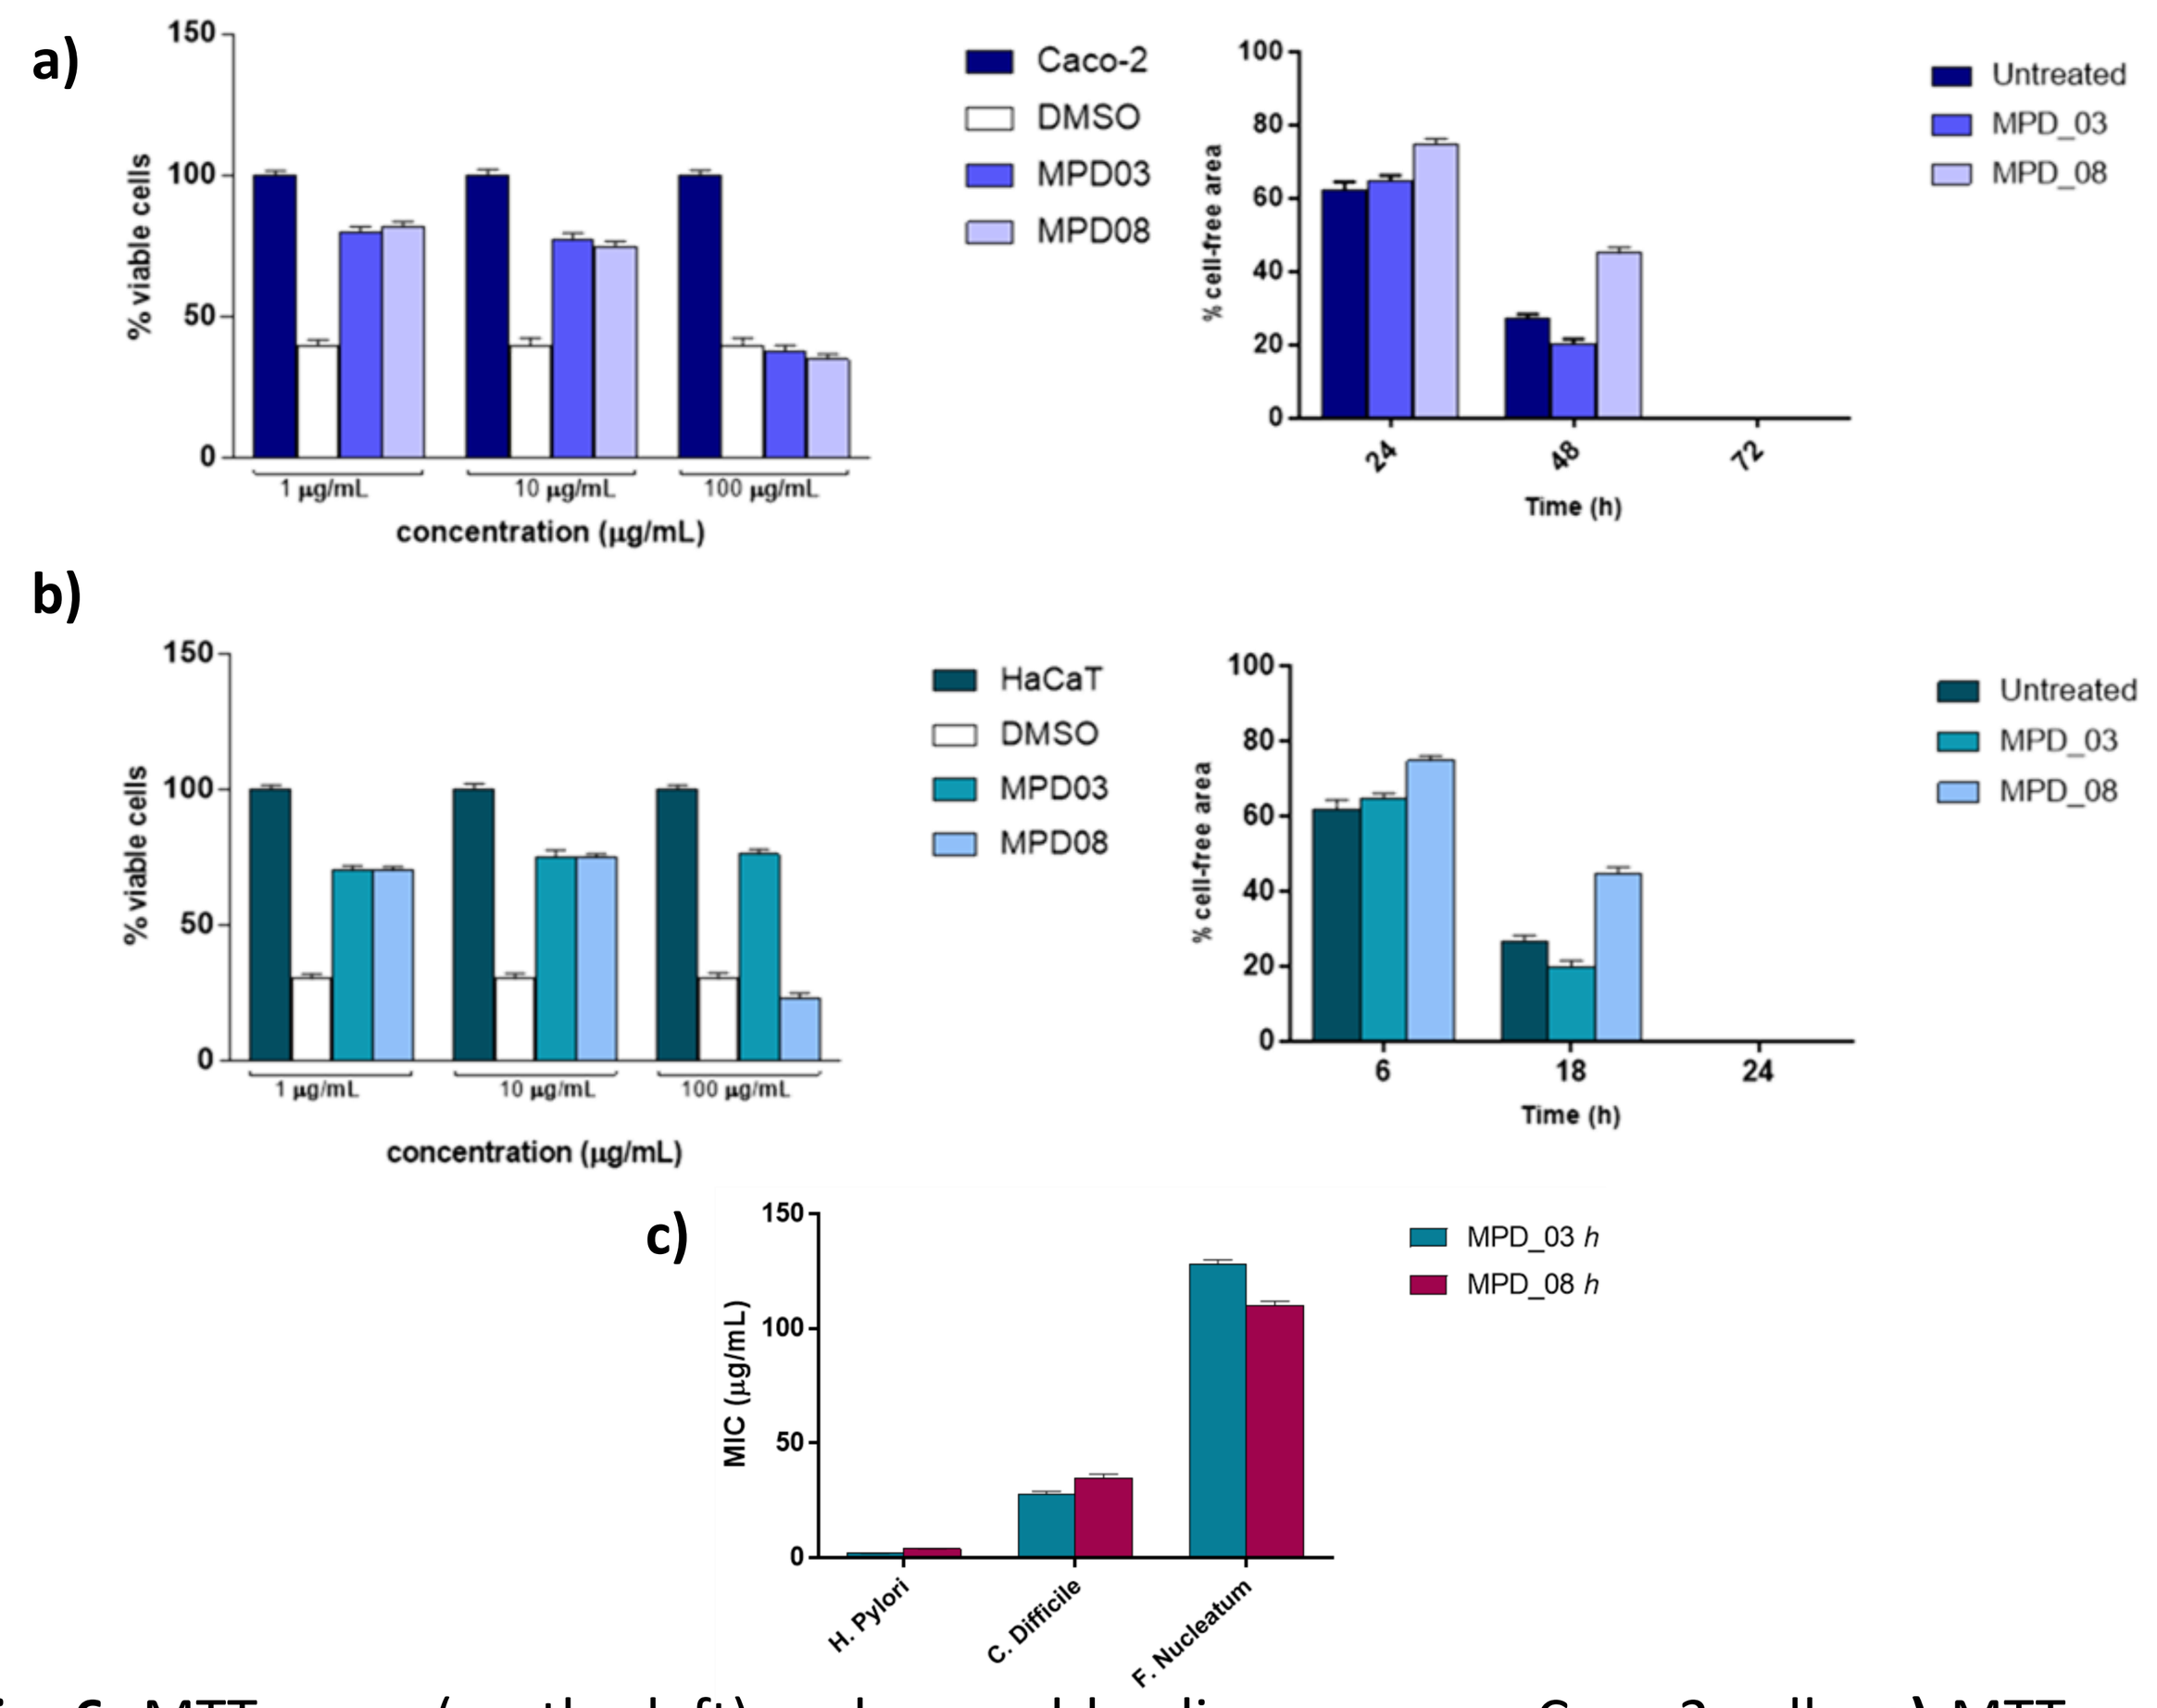

a)
Caco-2
DMSO
MPD03
MPD08
% viable cells
concentration (µg/mL)
Untreated
MPD_03
MPD_08
% cell-free area
Time (h)
b)
HaCaT
DMSO
MPD03
MPD08
c)
MIC (µg/mL)
MPD_03 h
MPD_08 h
H. Pylori
C. Difficile
F. Nucleatum

Fig. 6. MTT assay (on the left) and wound healing assay on Caco-2 cells; **a)** MTT assay (on the left) and wound healing assay on HaCaT cells; MIC values on *H. Pylori*, *C. Difficile,* and *F. Nucleatum* for **MPD_03** and **MPD_08.**

To determine the effect of **MPD_02-09** lipo-peptides, the MTT assays on Caco-2 **a**) and HaCaT **b**) cells were used. The results reported in **Fig. 6** panels **a)** and **b)** showed a % of cell viability comparable with those of untreated cells at 1 and 10 µg/mL concentrations. **MPD_02-09** showed wound dressing properties after 48h and 18h of incubation when assessed on Caco-2 and HaCat respectively.

Antimicrobial assays on *H. Pylori*, *C. Difficile,* and *F. Nucleatum* revealed MIC values ranging from 2 and 128 µg/mL.

FTIR ANALYSIS

Fourier Transform Infrared (FTIR) was used to examine the structure and the conformation of **MPD_03** and **MPD_08** lipo-peptides (**Fig. 7**).

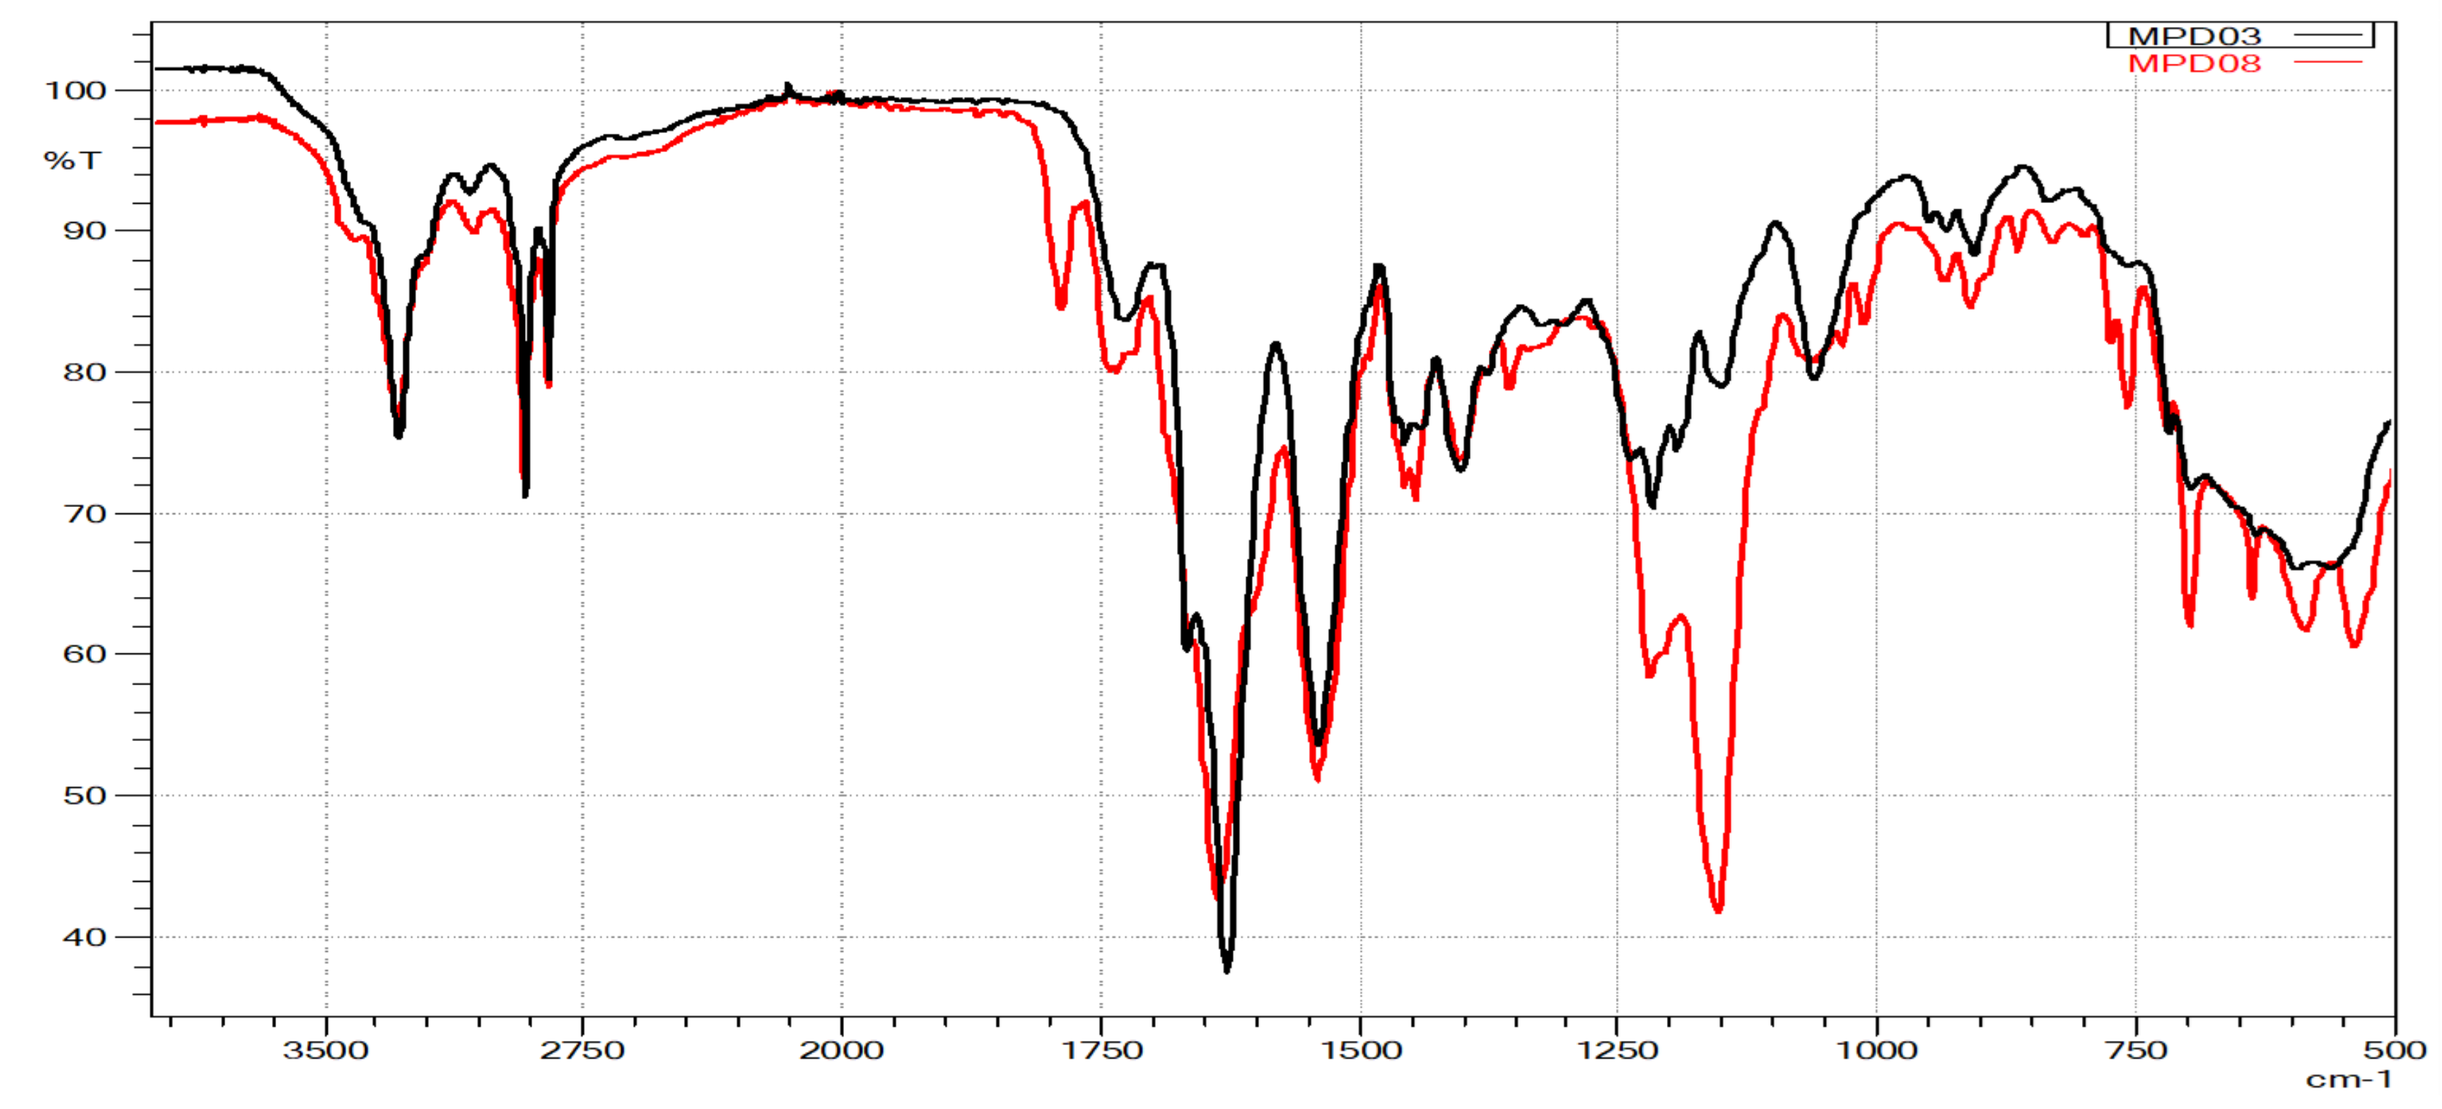

MPD03
MPD08
%T
cm-1

Fig. 7. FTIR of **MPD_03** (black line) and **MPD_08** (red line).

CONCLUSIONS AND FUTURE PERSPECTIVE

We proposed our **MPD*h*** platform as mucoadhesive gels designed for the controlled release of budesonide. This "sticky steroid" could represent a therapeutic option for eosinophilic esophagitis and inflammatory bowel diseases. The use of oral viscous media as delivery platforms of anti-inflammatory drugs, endorsed with antibacterial activity will be further evaluated *in vivo* animal model of gastrointestinal pathologies.

REFERENCES

1. Hong, J., et al. *Eur. Food Res. Technol.*, 2015, 240, 327-333.
2. Gubatan, J., et al. *World J. Gastroenterol*. 2021, 27, 7402-7422.
3. Li, Y., et al. Bioeng. *Transl. Med*. 2016, 1, 306-322.
4. Marinelli, L., et al. *Int. J. Pharm*. 2020, 30, 582.

SIMPOSIO AFI RIMINI 7 • 8 • 9 GIUGNO 2023
Associazione Farmaceutici Industria Società Scientifica
MUR Ministero dell'Università e della Ricerca
Finanziato dall'Unione europea NextGenerationEU